High prevalence of immune exclusion in cancer as determined by pathologist assessment and image analysis

Florent Peyraud[1], Fredrick D. Gootkind[2], Antoine Italiano[1], Alban Bessede[3], Jean-Philippe Guegan[3], Xinwei Sher[2], Thomas Schürpf[2], Guy T. Clifton[2], Laura A. Dillon[2]

[1]Early Phase Trials and Sarcoma Unit, Institut Bergonié, Bordeaux, France; University of Bordeaux, Bordeaux, France; [2]Parthenon Therapeutics, Boston, MA, USA; [3]Explicyte Immuno-Oncology, Bordeaux, France

2108

Background

- Immune infiltrated tumors have high levels of lymphocytes that penetrate the tumor parenchyma and come into contact with tumor cells.
- Tumors with few lymphocytes in contact with tumor cells can be divided into desert or excluded phenotypes based on lymphocyte absence/paucity or restriction to the peritumoral stroma, respectively.
- Distinguishing between immune infiltrated, desert, and excluded tumors is important since infiltrated tumors have been observed to be more responsive to checkpoint inhibitor therapy. However, standard methods to systematically identify and characterize immune phenotypes, particularly immune exclusion, for patient stratification are lacking.

Methods

- Slides from colorectal cancer (CRC), non-small cell lung cancer (NSCLC), ovarian cancer (OC), pancreatic ductal adenocarcinoma (PDAC), triple-negative breast cancer (TNBC), leiomyosarcoma (LMS), and undifferentiated pleomorphic sarcoma (UPS) were stained by multiplex IHC (mIHC) for CD8 and pan-cytokeratin (panCK).
- Pathologist assessment (PA) was done using the mIHC-stained slides to classify the tumors as: desert, with a paucity of CD8+ T cells; excluded, with CD8+ T cells not penetrating the tumor parenchyma; and infiltrated, with CD8+ T cells within the tumor parenchyma.
- For the carcinomas, adjacent sections were stained with a multiplex immunofluorescence (mIF) panel containing CD8 and a tumor cell marker. The tumor bed was identified by pathologist annotation.
- Image analysis (IA) was performed within the annotated tumor bed of the mIF images from CRC, NSCLC, OC, PDAC, and TNBC using QuPath. Representative samples from each indication were used to train a pixel-based image classifier that divided the samples into tumor epithelium (epithelium), healthy epithelium, stroma, and necrosis. DAPI-based cell detection was carried out and CD8+ cell density was quantified in the epithelium and stroma within the tumor bed.
- Samples were classified as desert, excluded, or infiltrated by IA using indication-specific cut-offs guided by the PA classification. The average CD8+ cell density and standard deviation in epithelium were used to calculate the cutoff between infiltrated and non-infiltrated (desert plus excluded). The average CD8+ cell density and standard deviation in stroma were used to calculate the cutoff between desert and excluded.

Disclosures

- Mr. Gootkind and Drs. Sher, Schürpf, Clifton, and Dillon are employees of Parthenon Therapeutics.
- Drs. Bessede and Guegan are employees of Explicyte.
- Dr. Italiano has research grants from AstraZeneca, Bayer, BMS, Chugai, Merck, MSD, Novartis, Pharmamar, and Roche. He serves on advisory boards. for AstraZeneca, Bayer, BMS, Chugai, Deciphera, Epizyme, Merck, MSD, Novartis, Parthenon Therapeutics, Pharmamar, and Roche.

Results

- Immune phenotypes were classified for 143 samples based on pathologist assessment of mIHC images and 103 samples by image analysis of mIF images (Table 1).
- Immune exclusion was observed in >50% of cases in TNBC, NSCLC, PDAC and CRC by pathologist assessment and in PDAC and CRC by image analysis. Image analysis differed from pathologist assessment in 25 (24.3%) cases (Figure 1).

Table 1.		Pathologist Assessment Classification			Image Analysis Classification		
Tumor Type	n	Desert (%)	Excluded (%)	Infiltrated (%)	Desert (%)	Excluded (%)	Infiltrated (%)
CRC	20	7 (35.0)	10 (50.0)	3 (15.0)	6 (30.0)	11 (55.0)	3 (15.0)
NSCLC	21	5 (23.8)	13 (61.9)	3 (14.3)	6 (28.6)	9 (42.9)	6 (28.6)
OC	20	5 (25.0)	7 (35.0)	8 (40.0)	9 (45.0)	3 (15.0)	8 (40.0)
PDAC	21	7 (33.3)	13 (61.9)	1 (4.8)	7 (33.3)	12 (57.1)	2 (9.5)
TNBC	21	4 (19.0)	14 (66.6)	3 (14.3)	4 (19.0)	10 (47.6)	7 (33.3)
LMS	20	6 (30.0)	1 (5.0)	13 (65.0)	NA*	NA*	NA*
UPS	20	4 (20.0)	1 (5.0)	15 (75.0)	NA*	NA*	NA*
Total	143/103	38 (26.6)	59 (41.3)	46 (32.1)	32 (31.1)	45 (43.7)	26 (25.2)

*NA = not analyzed

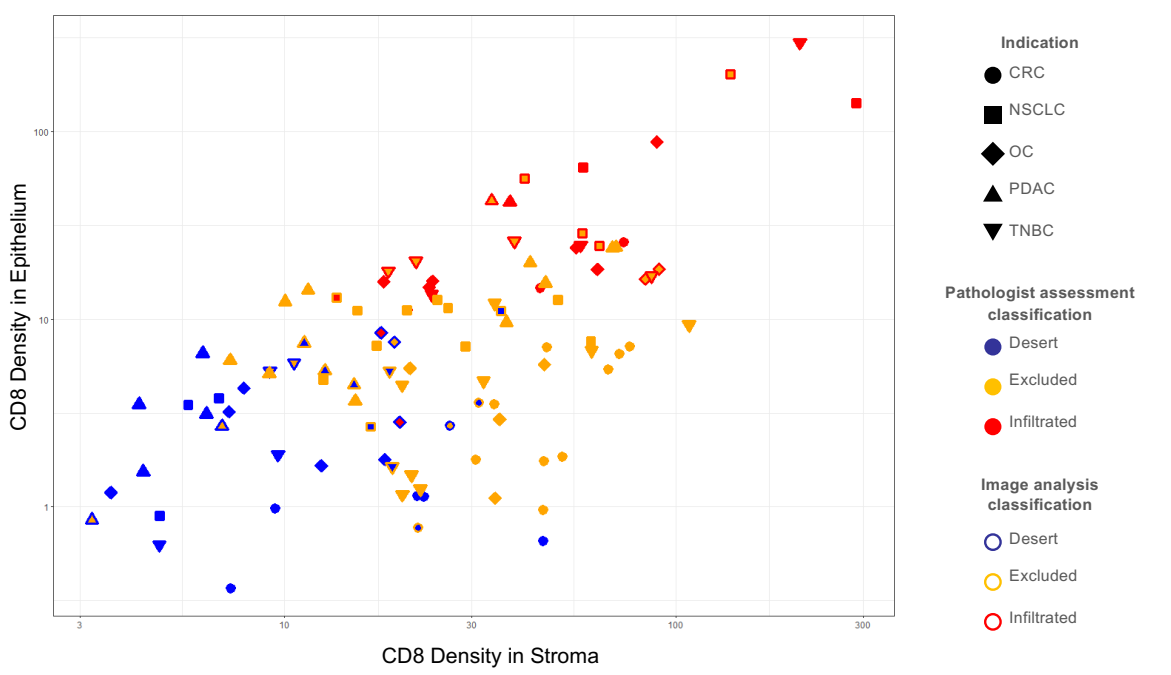
CD8 Density in Epithelium
CD8 Density in Stroma
100
10
1
3
10
30
100
300
Indication
CRC
NSCLC
OC
PDAC
TNBC
Pathologist assessment classification
Desert
Excluded
Infiltrated
Image analysis classification
Desert
Excluded
Infiltrated

Figure 1. Immune classification of the carcinomas. Indication is shown by shape, pathologist assessment classification by data point fill color, and image analysis classification by data point border color. Images with different fill and border colors were discordant between PA and IA classification.

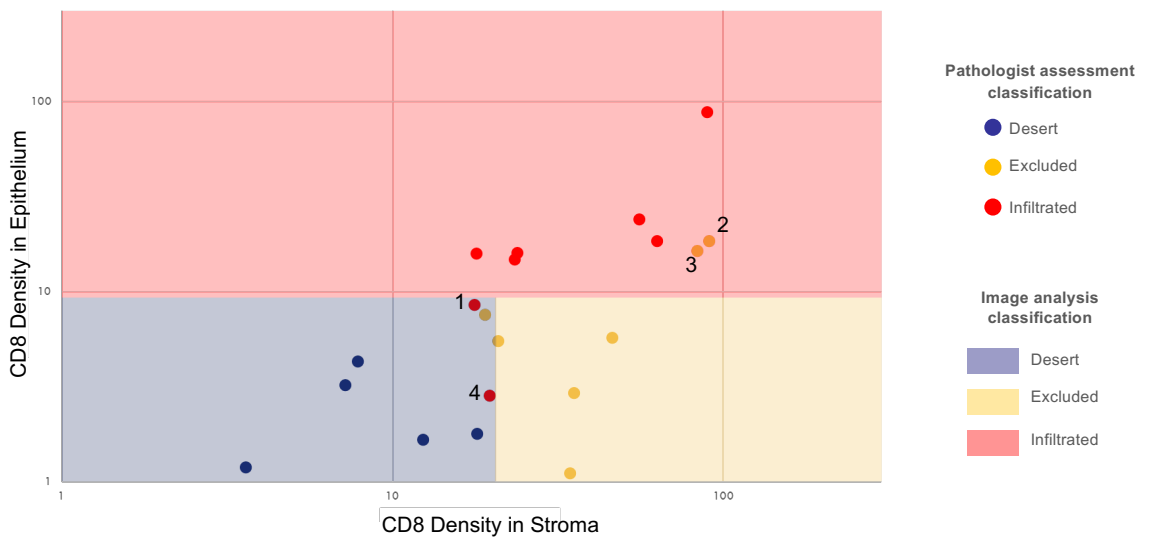
CD8 Density in Epithelium
CD8 Density in Stroma
100
10
1
1
10
100
1
2
3
4
Pathologist assessment classification
Desert
Excluded
Infiltrated
Image analysis classification
Desert
Excluded
Infiltrated

Figure 2. Immune classification of the ovarian cancer samples. Pathologist assessment classification is shown by the fill color of each data point and image analysis classification by plot background color. Examples shown at right are numbered 1-4.

- Pathologist review of the discordant cases revealed that discrepancies were generally due to tumor heterogeneity, thresholding, assessment of cells at the epithelium-stroma boundaries, necrosis, and artifacts. Examples of discordant cases within the ovarian cancer samples are shown below and annotated in Figure 2.

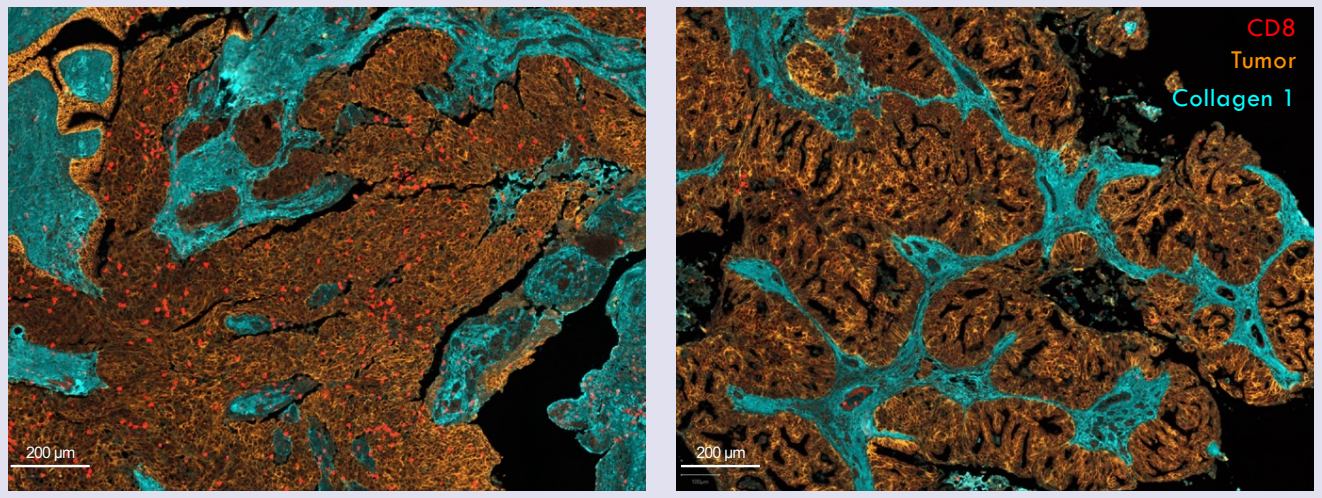
CD8
Tumor
Collagen 1
200 µm
200 µm

Example 1: Intra-slide heterogeneity of CD8+ cell density contributes to discordant classification of this ovarian cancer sample as "infiltrated" by pathologist assessment and "desert" by image analysis.

Example 2: This ovarian cancer sample has high CD8+ cell density in epithelium but higher CD8+ cell density in stroma. Lack of firm thresholds regarding how CD8+ cell density in epithelium (outright or as compared to CD8+ cell density in stroma) should relate to immune phenotype classification lead to this sample being classified as "excluded" by pathologist assessment and "infiltrated" by image analysis.

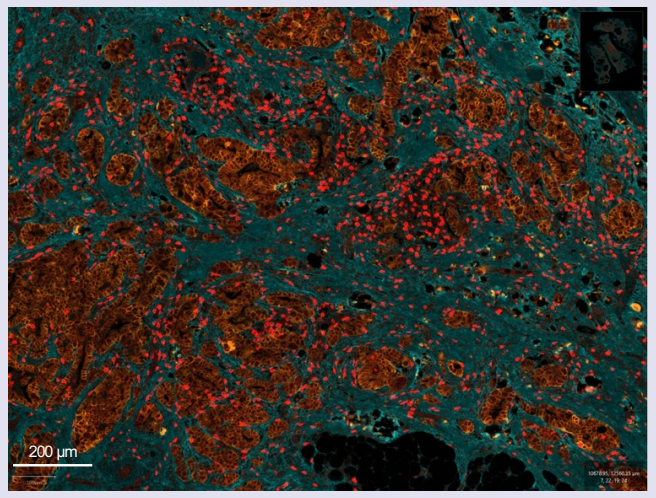
200 µm

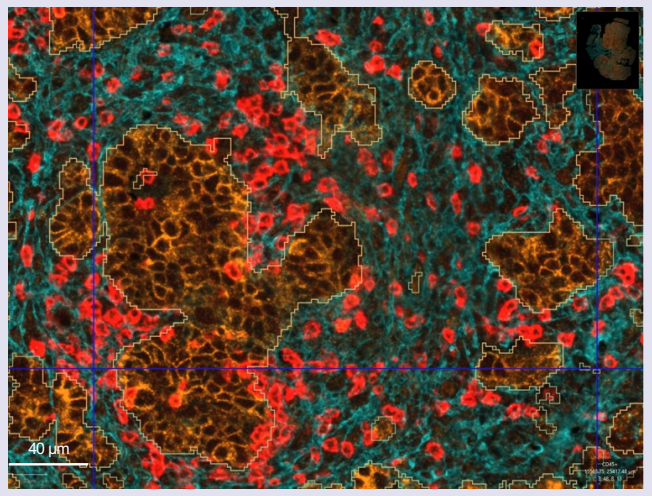
40 µm

Example 3: Significant numbers of CD8+ cells in this ovarian cancer sample are found at the boundaries between epithelium and stroma, lending ambiguity as to their location. This sample was classified as "excluded" by pathologist assessment and "infiltrated" by image analysis.

Example 4: Image analysis was unable to reliably detect necrosis in ovarian cancer sample 4 (necrotic area detected as stroma), reducing the reliability of quantification of CD8+ cell density in the tissue. This sample was classified as "infiltrated" by pathologist assessment and "desert" by image analysis.

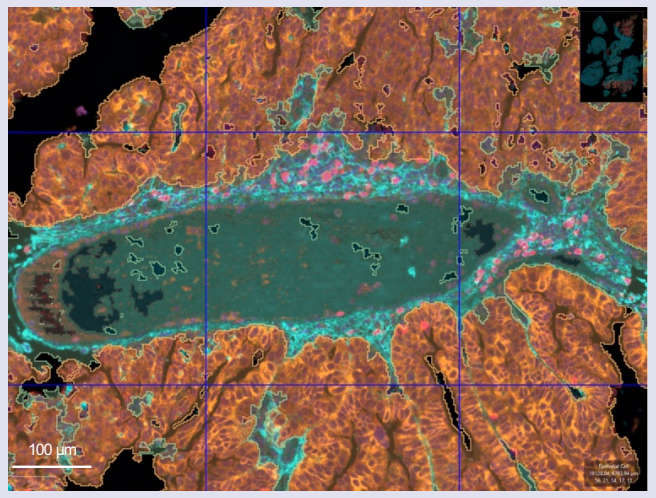
100 µm

Conclusions

- Immune exclusion is highly prevalent in the examined carcinoma types as determined by pathologist assessment and image analysis, but lower in sarcoma, where most samples were classified as infiltrated by pathologist assessment.
- Image analysis-based approaches, guided by pathologist input, offer promise to quantitatively determine tumor immune phenotypes in a quick and systematic way to guide patients to the most effective therapy.

Bergonié UNICANCER NOUVELLE-AQUITAINE

PARTHENON THERAPEUTICS